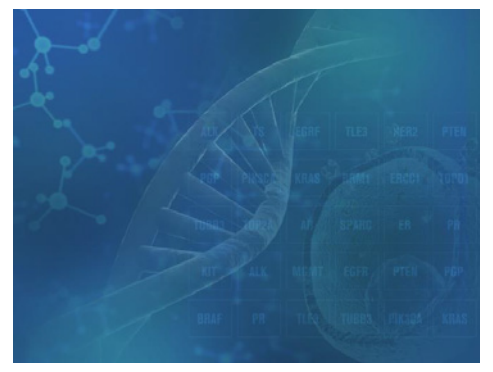

Comparison of Mutations and Protein Expression in Potentially Actionable Targets in 5500 Triple Negative vs. non-Triple Negative Breast Cancers

Joyce A O'Shaughnessy[1], Zoran Gatalica[2], Jeffery Kimbrough[2], Sherri Z Millis[2]

[1]Baylor Sammons Cancer Center, Texas Oncology, US Oncology, Dallas TX, [2]Caris Life Sciences, Phoenix, AZ

San Antonio Breast Cancer Symposium – Cancer Therapy and Research Center at UT Health Science Center - December 10-14, 2013

Introduction

Triple negative breast cancer is a heterogeneous disease with no established targeted treatment options for patients with metastatic disease. This study was undertaken to profile a large commercial biomarker database in an effort to identify potential molecular differences between triple negative and non-triple negative breast cancers and to identify potential new molecular therapeutic targets.

Methods

A cohort of 5521 patient samples (profiled at Caris Life Sciences between 2009 and Sep. 2013 generally from patients with metastatic disease) was evaluated for similarities and differences in gene mutation (Sanger or Illumina), protein expression (immunohistochemistry), and/or gene amplification (CISH or FISH) between triple negative and non-triple negative breast cancers. The cohort was grouped by ER, PR, and Her2 IHC status (Figure 1).

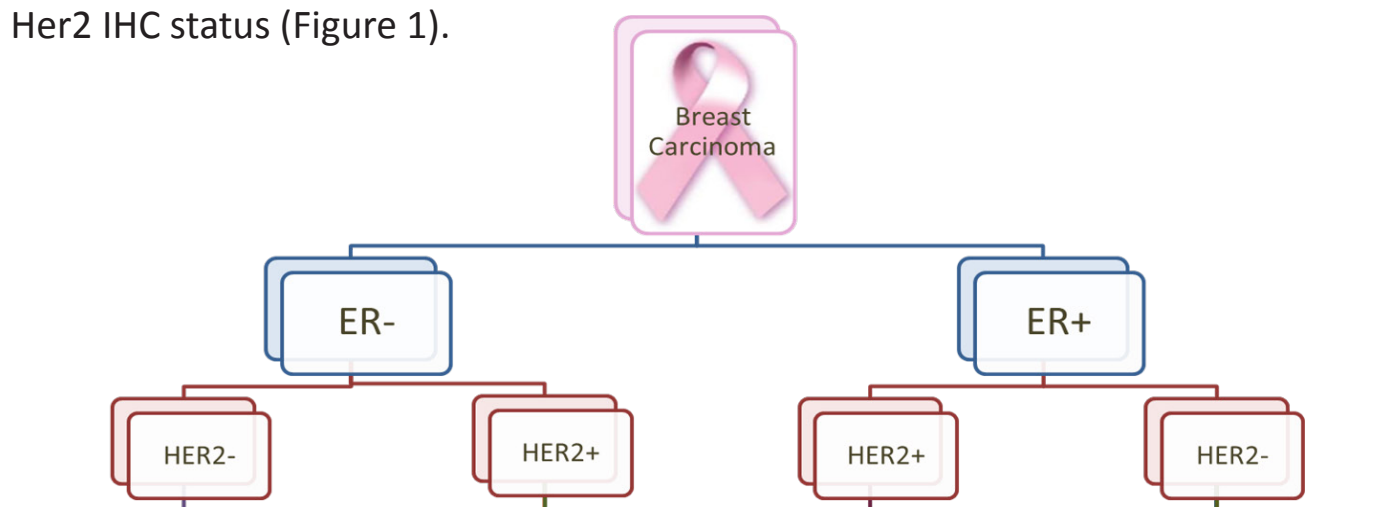
Breast Carcinoma
ER-
ER+
HER2-
HER2+
HER2+
HER2-

	PR- (1975)	PR+ (125)	PR+ (33)	PR- (310)	PR+ (133)	PR- (125)	PR+ (1867)	PR- (924)
1° vs Met	968 v 1007	62 v 63	12 v 21	133 v 177	81 v 52	53 v 72	907 v 960	281 v 641
Med. Age	53	55	55	56	51	57	57	58

Figure 1. Categorization of breast carcinomas based on ER, PR, Her2 status by IHC. Median age of each group and primary versus metastatic disease status is indicated below each category. *Each group is color coded for coordination throughout the poster.*

The samples were stained with the appropriate antibody to determine hormone receptor status, and the distribution of molecular subtypes was determined.
ER and PR was positive when 1% or more tumor cells nuclei stained with any intensity (graded as 1 to 3+). Her2 was positive when >10% of cells exhibited strong complete membranous staining (3+).

35.8% of the cases were TNBC. Due to the aggressive nature of TNBC, a higher percentage of TNBC patients is evaluated for molecular profiling than the general breast cancer population.

52.8% of the cohort was either ER or PR positive and HER2-. 10.9% of the patient cohort was HER2+, and in that cohort, 2.4% was positive for ER, PR and HER2 (Figure 2).

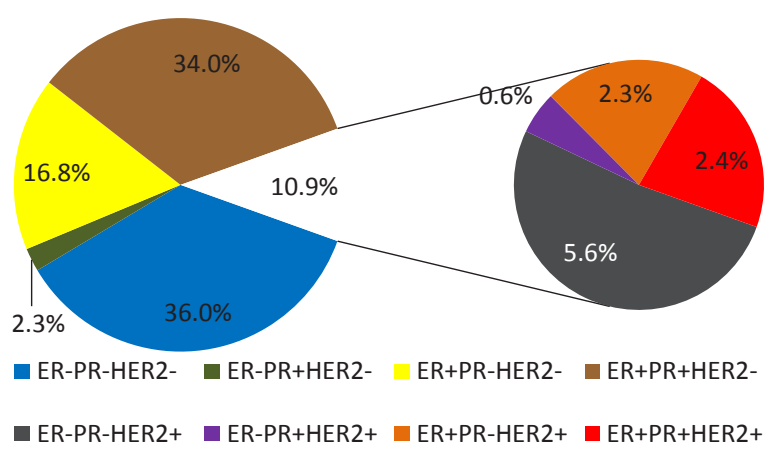
34.0%
16.8%
10.9%
2.3%
36.0%
0.6%
2.3%
2.4%
5.6%
ER-PR-HER2-
ER-PR+HER2-
ER+PR-HER2-
ER+PR+HER2-
ER-PR-HER2+
ER-PR+HER2+
ER+PR-HER2+
ER+PR+HER2+

Figure 2. Percent distribution by subtype.

Results: Immunohistochemistry (IHC) (% PTS +)

Case Total	Cancer Subtype	AR	c-kit	ERCC1	Ki67	MGMT*	PGP	PTEN*	RRM1	SPARC	TLE3	TOP2A	TOPO1	TS	TUBB3*
133	ER+PR+HER2+	81.4	1.2	65.1	73.7	68.3	3.9	56.9	32.3	55.3	72.3	64.4	73.0	10.0	26.7
125	ER+PR-HER2+	63.5	1.3	60.3	80.3	66.7	5.1	50.0	39.6	46.6	59.8	59.0	70.3	11.1	52.6
1867	ER+PR+HER2-	76.5	4.3	55.4	50.8	66.0	6.0	45.2	25.4	50.7	67.0	41.7	72.3	9.2	28.8
924	ER+PR-HER2-	59.1	6.1	45.7	55.9	69.2	10.6	43.1	29.1	48.0	59.2	38.8	72.8	9.6	35.4
33	ER-PR+HER2+	48.1	0.0	81.3	75.0	50.0	0.0	58.1	42.9	51.7	50.0	75.0	60.0	13.0	66.7
310	ER-PR-HER2+	50.5	4.9	46.0	84.3	52.9	12.0	37.3	33.2	51.5	52.8	60.8	72.1	16.5	47.7
125	ER-PR+HER2-	18.9	29.4	64.2	83.0	67.4	10.8	33.9	46.3	49.1	40.4	61.4	74.1	28.6	50.0
1975	ER-PR-HER2-	17.5	25.9	42.1	85.2	58.9	12.0	30.6	33.7	44.9	34.2	66.7	70.2	20.6	51.2

Table 1. IHC results expressed as percent positive cases (thresholds below). Grayed cells indicate < 50 cases tested.
*Expression of the biomarker below the threshold is considered predictive of response to therapy.

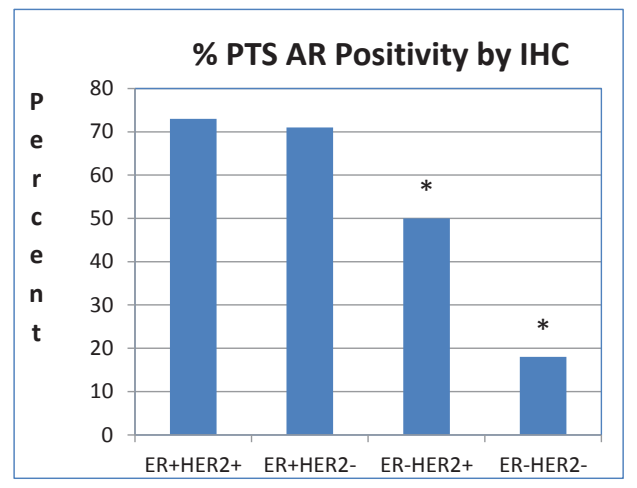
% PTS AR Positivity by IHC
Percent
80
70
60
50
40
30
20
10
0
*
*
ER+HER2+
ER+HER2-
ER-HER2+
ER-HER2-

Figure 3. AR expression levels by IHC. Significantly (p<0.05) lower expression of AR was seen in ER- negative tumors and further negatively affected by Her2- status (in ER- cases).

Table 2. Thresholds for IHC Biomarkers
AR =0+ or <10% or ≥1+ and ≥10%
cKIT =0+ and =100% or ≥2+ and ≥30%
cMET = <50% or <2+ or ≥2+ and ≥50%
ERCC1 =2+ and <50% or ≥3+ and ≥10%
Ki67 = ≥ 20%
MGMT =0+ or ≤35% or ≥1+ and >35%
PGP =0+ or <10% or ≥1+ and ≥10%
PTEN =0+ or ≤50% or ≥1+ and >50%
RRM1 ==0+ or <50% or <2+ or ≥2+ and ≥50%
SPARC =<30% or <2+ or ≥2+ and ≥30%
TLE3 =<30% or <2+ or ≥2+ and ≥30%
TOP2A =0+ or <10% or ≥1+ and ≥10%
TOPO1 =0+ or <30% or <2+ or ≥2+ and ≥30%
TS =0+ or ≤3+ and <10% or ≥1+ and ≥10%
TUBB3 =<30% or <2+ or ≥2+ and ≥30%

Results, In Situ Hybridization

Case Totals*	Cancer Subtype	cMET	cMYC	EGFR	HER2	TOP2A
100	ER+PR+HER2+	0	28.4	16.4	90.5	33.0
100	ER+PR-HER2+	4.2	20.8	7.1	93.2	38.8
700	ER+PR+HER2-	1.5	10.4	8.3	5.0	6.2
300	ER+PR-HER2-	3.0	14.9	9.5	6.6	6.1
175	ER-PR-HER2+	5.4	25.9	25.4	94.1	16.5
600	ER-PR-HER2-	1.6	22.1	21.7	4.6	3.7

Table 3. ISH results expressed as percent cases positive for gene amplification. Grayed cells indicate <50 cases tested. *Case totals are averaged, as not all cases had all tests performed.

HER2 FISH: HER2/neu:CEP 17 signal ratio of >=2.0 is amplified and <2.0 is not amplified; 1.8-2.2 is equivocal.
cMET CISH: >= 5 copies is amplified
TOP2A:CEP17 signal ratio of >=2.0 is amplified
EGFR: ≥ 4 copies in ≥ 40% of tumor cells.

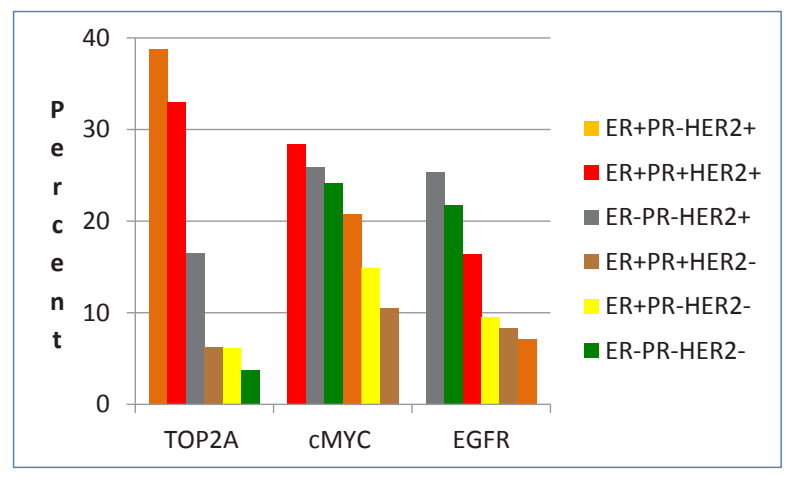
Percent
40
30
20
10
0
TOP2A
cMYC
EGFR
ER+PR-HER2+
ER+PR+HER2+
ER-PR-HER2+
ER+PR+HER2-
ER+PR-HER2-
ER-PR-HER2-

Figure 4. ISH Results for 3 genes with significantly different amplification, distributed from highest to lowest by category.

Results: Sequencing (% PTS with Mutations)

A. Cancer Subtype	ABL1	AKT1	APC	ATM	BRAF	CDH1	c-kit	cMET	EGFR	ERBB2	ERBB4	KRAS	PIK3CA	PTEN	RB1	STK11	TP53
ER+HER2+ PR+/-	0.0	0.0	6.5	3.2	0.0	3.2	0.0	0.0	0.0	9.7	3.2	0.0	29.8	3.2	0.0	0.0	37.9
ER+HER2- PR+/-	1.2	3.7	4.6	2.3	0.5	0.6	1.8	1.1	0.8	1.2	0.6	0.9	37.6	4.9	1.4	2.1	28.1
ER-HER2+ PR+/-	0.0	0.0	5.1	0.0	0.0	0.0	1.7	2.6	0.0	2.6	0.0	2.4	36.9	2.6	0.0	0.0	78.4
ER-HER2- PR-	0.4	3.3	4.0	0.4	0.5	0.0	0.8	2.2	1.0	2.6	0.4	1.6	14.6	6.3	1.8	0.8	63.7

Table 4. A. Sequencing results (Sanger or NGS) expressed as percent positive cases with mutations. Grayed cells indicate < 50 cases tested.
B. Total cases tested by each technology.

B. Cancer Subtype	Case Total by NGS	Case Total ,Sanger (BRAF, c-kit, KRAS, PIK3CA)
ER+HER2+ PR+/-	31	~50
ER+HER2-PR+/-	350	~350
ER-HER2+ PR+/-	40	~60
ER-HER2- PR-	275	~250

Figure 5. Alteration frequency of PIK3CA and TP53.

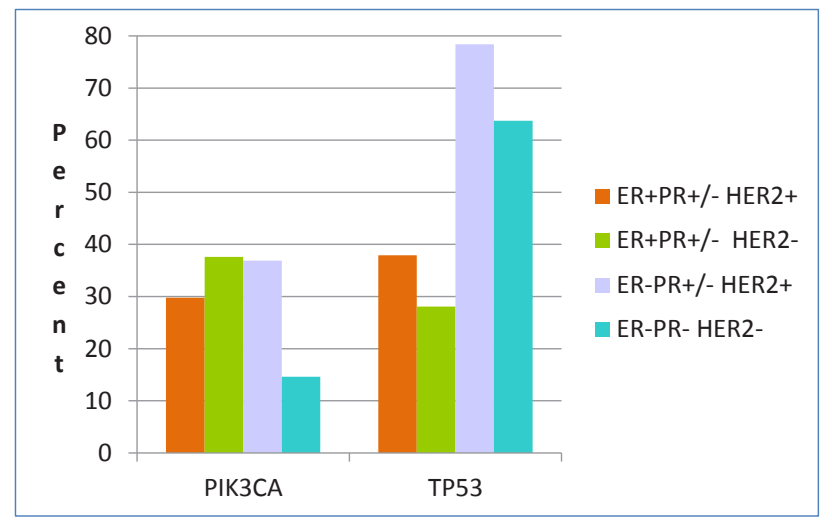
Percent
80
70
60
50
40
30
20
10
0
PIK3CA
TP53
ER+PR+/- HER2+
ER+PR+/- HER2-
ER-PR+/- HER2+
ER-PR- HER2-

TNBC patients had a significantly lower PIK3CA mutation rate than all other subtypes (p<0.05) and a significantly higher TP53 mutation rate than the receptor positive cases (p<0.05) . In fact, TP53 is significantly more commonly mutated in ER- tumors, *irrespective* of HER2 status. Additionally, ERBB2 mutations are seen in all subtypes.

Results: ISH and Sequencing Concordance

The cases were analyzed for both HER2 gene amplification and HER2 mutation. 1 of 18 ER+PR-HER2+ cases, 1 of 228 ER+PR+HER2- by IHC cases, and 2 of 271 TNBC by IHC cases assayed were positive for both HER2 gene amplification and a HER2 mutation.

Results: PIK3CA/mTOR Pathway Alterations in AR+ PTS

Cancer Subtype	Total cases AR+ and PIK3CA assayed	Percent Cases with PIK3CA mutation	Total cases with AR+ and PTEN assayed	Percent Cases with PTEN loss (IHC) or mutation	Percent Cases with both PIK3CA mutation/ PTEN loss or mutation
ER+HER2-	499	39%	1811	0.4% PTEN mut 53.5% PTEN loss 53.9% Total	8%
ER+HER2+	117	26%	167	0.6% PTEN mut 40.1% Pten loss 40.7% Total	12%
ER-HER2+	102	38%	160	0.6% PTEN mut 55.7% PTEN loss 56.3% Total	20%
ER-HER2-	75	29%	339	1.5% PTEN mut 60.4% PTEN loss 61.9% Total	11%

Table 5. PIK3CA and/or PTEN status in AR positive (IHC) cases. No genomic differences were seen between primary and metastatic cases, with the exception of the ER+HER2+ subtype, where there was almost a two-fold increase in PIK3CA(18% vs 34%), PTEN (26% vs 47%), or both (5% vs 19%) mutations in primary vs metastatic cases (p<0.05).

Results: AR/Ki67 Relationships

A. Cancer Subtype	AR expression (IHC)	ki67 index Low (<15%)	ki67 index High (>=15%)	# Cases
ER+ HER2-	AR+	50.0%	50.0%	1019
	AR-	49.7%	50.3%	342
ER+ HER2+	AR+	23.3%	75.7%	103
	AR-	23.1%	76.9%	26

B. Cancer Subtype	AR expression (IHC)	ki67 index (<30%)	ki67 index (>=30%<60%)	ki67 index (>=60%)	# Cases
ER- HER2-	AR+	47%	33%	20%	201
	AR-	21%	30%	49%	841
ER- HER2+	AR+	38%	38%	24%	94
	AR-	31%	36%	33%	88

Table 6A, B. Relationship between AR status and Ki67 index for *A.* ER positive and *B.* ER negative breast cancers.

Conclusions

- **AR is expressed in 50% of ER- HER2+ and 18% of triple negative breast cancers and may be an important therapeutic target.**
- **Nearly all AR+ cases have PIK3CA mutation or PTEN loss/mutation suggesting PI3K pathway activation. Combined AR and PI3K inhibition should be evaluated.**
- **In TNBC but not ER+ or HER2+ disease, AR expression is associated with decreased proliferation.**
- **In these poor prognosis ER+ cancers, nearly all had evidence of PI3K pathway activation and about 30% had p53 mutations.**
- **Outside of p53 and PIK3CA, targetable, activating mutations occur with low frequency across breast cancer subtypes.**
- **APC mutations occur in 5% of breast cancers across subtypes and whether these may predict for benefit from anti-frizzled receptor therapy should be explored.**
- **EGFR gene amplification occurs in about 10% of poor prognosis ER+ and 20% of ER- breast cancers. Whether this finding predicts for benefit from anti-EGFR therapy is worthy of investigation.**
- **Multi-platform molecular profiling is needed to identify targetable genomic and proteomic alterations in poor prognosis breast cancer.**

References

1. Gucalp, A., et al. Phase II trial of bicalutamide in patients with androgen receptor-positive, estrogen receptor-negative metastatic Breast Cancer. Clin Cancer Res. 2013 Oct 1;19 (19):5505-12.
2. The Cancer Genome Atlas Network, Comprehensive molecular portraits of human breast tumours, Nature 490,61–70, 2012
3. Zhen Wang, Targeting p53 for Novel Anticancer Therapy, Transl Oncol. 2010 February; 3(1): 1–12.

This presentation is the intellectual property of the author/presenter. Contact bstengle@carisls.com for permission to reprint and/or distribute.